THE APPLICATION OF PHYSICOCHEMICAL METHODS OF ANALYSIS TO THE DEVELOPMENT OF CONJUGATE VACCINES AGAINST MENINGOCOCCAL GROUP A BACTERIA

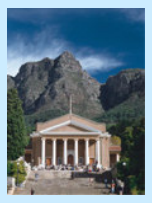

Neil Ravenscroft[1], Meredith Hearshaw[1], Priscilla Mensah[1], Suresh Beri[2], Jayant Joshi[2], Karupothula Suresh[2], Akshay Goel[2], Jean-Marie Preaud[3], Simonetta Viviani[3], Marc LaForce[3]

[1]Department of Chemistry, University of Cape Town, Cape Town, South Africa [2]Serum Institute of India Limited, Pune, India
[3]Meningitis Vaccine Project (WHO/PATH), Ferney-Voltaire, France

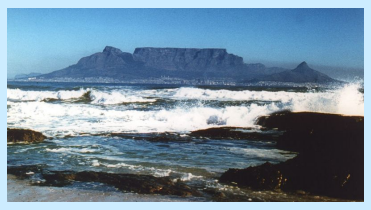

1. INTRODUCTION

The meningococcal group A organism is responsible for 90% of the cases of endemic and epidemic meningitis caused by *Neisseria meningitidis* bacteria. Preventive immunisation should avoid a great number of deaths and be less expensive than mass immunisation campaigns performed after epidemics have begun. This is best achieved by vaccination with conjugate vaccines which, unlike polysaccharide vaccines, are immunogenic in the very young, induce immunological memory and are likely to give long-lasting protection. MVP (Meningitis Vaccine Project [1]) is developing an affordable monovalent meningococcal A conjugate vaccine for sub-Saharan Africa which is manufactured by Serum Institute of India Limited (SIIL) using aldehyde-hydrazide condensation chemistry (panel 2) developed at the US Center for Biologics Evaluation and Research [2]. Manufacturers and regulatory authorities have had decades of experience with meningococcal capsular polysaccharide vaccines, but the production and control of conjugate vaccines are more complex and vaccine specific. Successful development of the conjugate vaccine requires control of the production process from starting polysaccharide, to the formation of activated intermediates and their conjugation to yield the conjugate vaccine, as well as demonstration of manufacturing consistency. Since group A meningococcal conjugate vaccines are manufactured from purified components by a clearly defined chemical process, the strategy for the control of the vaccine relies heavily on determination of the molecular characterization and purity of each vaccine lot and intermediates [3]. Recent advances in bioanalytical methodology [4] permit the detailed structural characterization of conjugate vaccines and intermediates to be achieved by the use of physicochemical techniques. including chromatography (panel 3) and nuclear magnetic resonance spectroscopy (panel 4).

2. CONJUGATION PROCESS and CONTROL

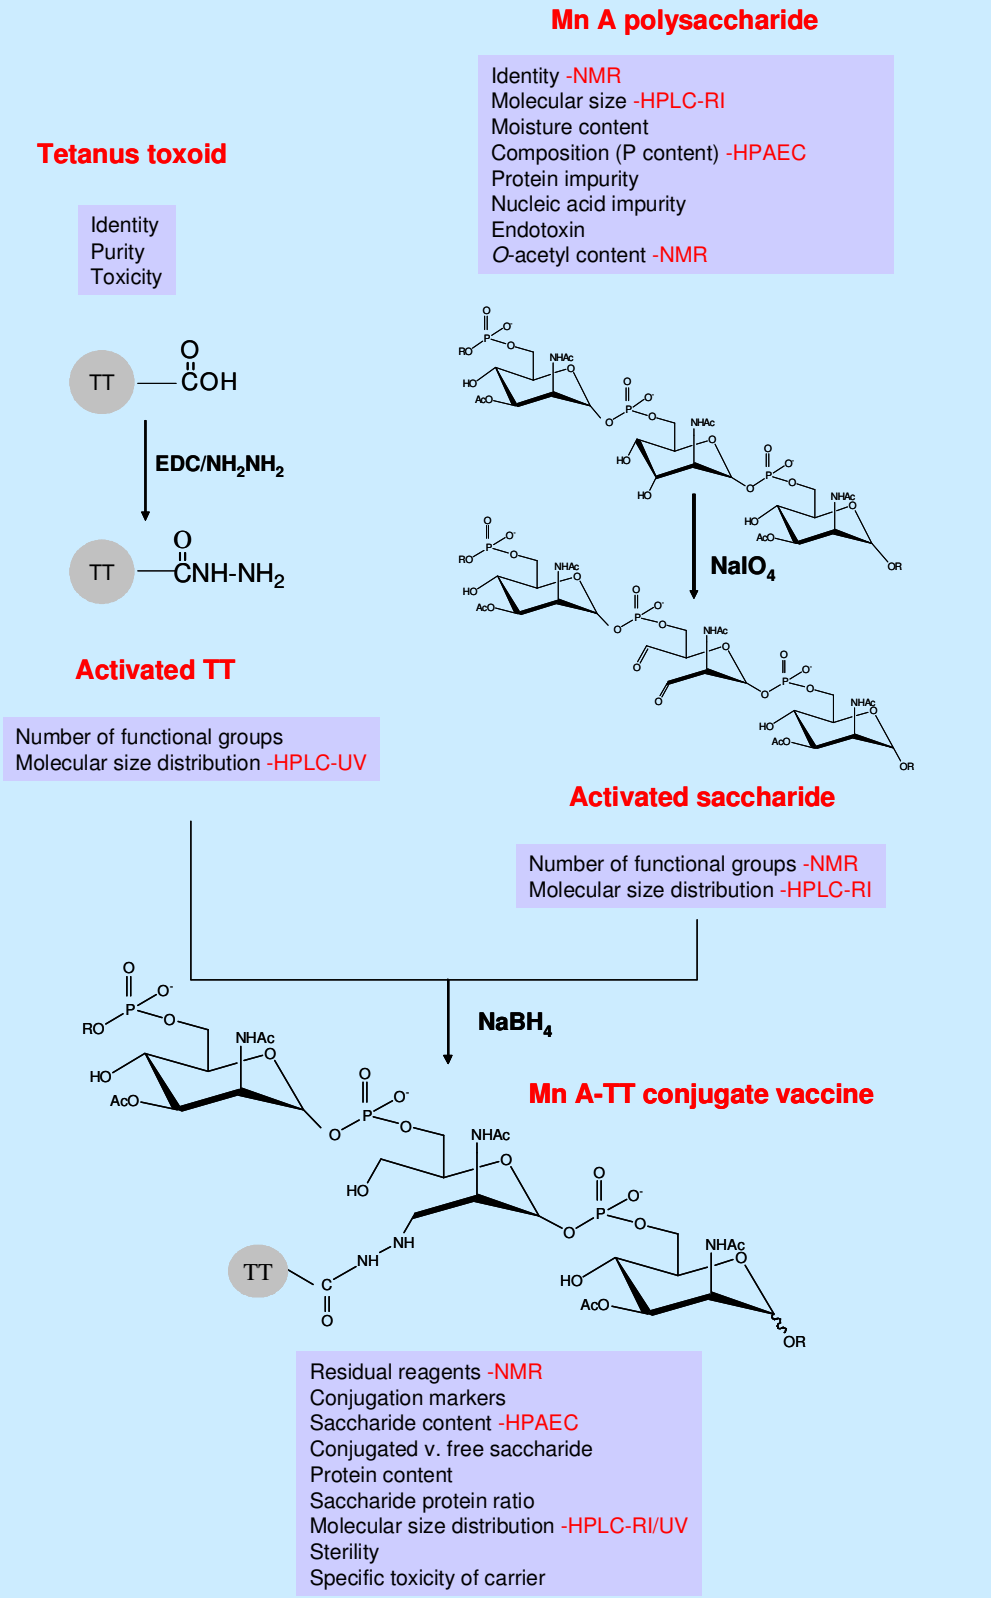

Mn A polysaccharide
Identity -NMR
Molecular size -HPLC-RI
Moisture content
Composition (P content) -HPAEC
Protein impurity
Nucleic acid impurity
Endotoxin
O-acetyl content -NMR
Tetanus toxoid
Identity
Purity
Toxicity
TT
COH
EDC/NH2NH2
CNH-NH2
Activated TT
Number of functional groups
Molecular size distribution -HPLC-UV
NaIO4
Activated saccharide
Number of functional groups -NMR
Molecular size distribution -HPLC-RI
NaBH4
Mn A-TT conjugate vaccine
Residual reagents -NMR
Conjugation markers
Saccharide content -HPAEC
Conjugated v. free saccharide
Protein content
Saccharide protein ratio
Molecular size distribution -HPLC-RI/UV
Sterility
Specific toxicity of carrier

3. CHROMATOGRAPHY

High-performance anion-exchange chromatography with pulsed amperomeric detection (HPAEC-PAD) was performed on a Dionex DX-500 system fitted with a GP50 pump, ED50 detector and AS50 autosampler, using a CarboPac PA1 column and a NaOH/Na acetate elution gradient. High performance size-exclusion chromatography (SEC-HPLC) was performed on a Waters instrument with ultra-violet (UV) or refractive index (RI) detection.

HPAEC-PAD: Mn A saccharide content

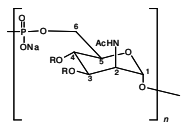

Colorimetric assays for P
simple but poor selectivity and sensitivity-no int. std.

New method: Acid hydrolysis (TFA) and HPAEC-PAD analysis of Monomer 6-P-mannosamine [5]

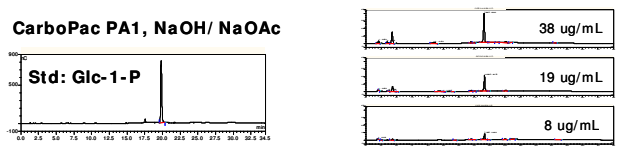

CarboPac PA1, NaOH/NaOAc
Std: Glc-1-P
38 ug/mL
19 ug/mL
8 ug/mL

- **Applicable to Mn A PS, activated saccharide and Mn A-TT conjugate**

SEC-HPLC-RI: Mn A saccharide size analysis

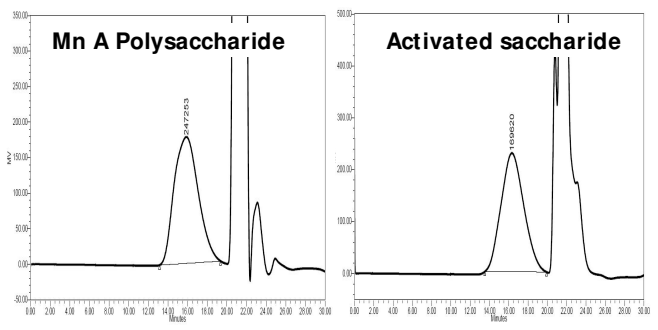

Mn A Polysaccharide
Activated saccharide

- **Size profiles of Mn A polysaccharide lots & activated saccharide**
- **Track Mn A PS activation reaction**

SEC-HPLC-UV/RI: TT and Mn A-TT conjugate

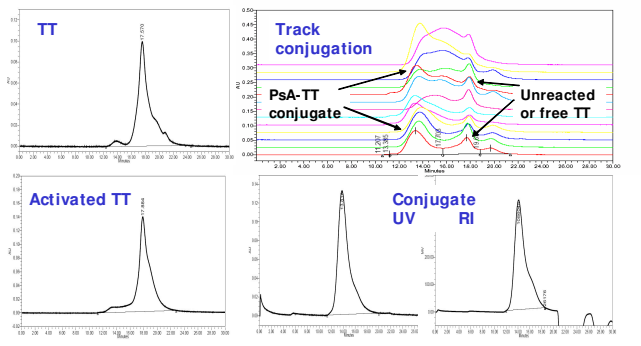

TT
Track conjugation
PsA-TT conjugate
Unreacted or free TT
Activated TT
Conjugate
UV
RI

- **Size profiles of TT, activated TT and Mn A-TT conjugate**
- **Track TT activation and conjugation reaction**

Total and free saccharide by HPAEC-PAD

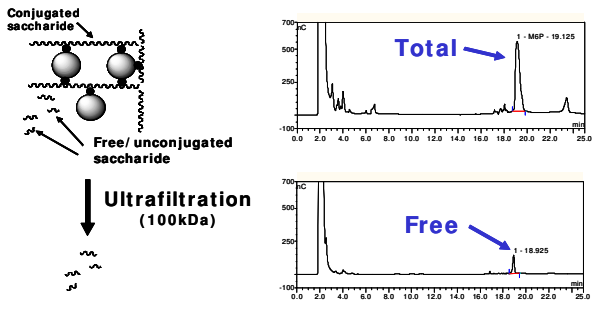

Conjugated saccharide
Free/unconjugated saccharide
Ultrafiltration
(100kDa)
Total
1 - M6P - 19.125
Free
1 - 18.925

Conjugate bulk: ± 10% free saccharide after one year at 4ºC

- **Integrity of Mn A-TT conjugate shown by free saccharide assay**

4. SPECTROSCOPY

NMR spectra were acquired on a Varian Unity 400 (University of Cape Town) or 500 (NIBSC) NMR spectrometer. Samples of saccharide or conjugate (2-10mg) were freeze-dried and exchanged three times with D_2O prior to analysis, which was performed in D_2O at 300K. De-Oacetylated samples were prepared by addition of 200mM NaOD to the sample in the NMR tube.

¹H-NMR: Mn A PS structure/identity-1

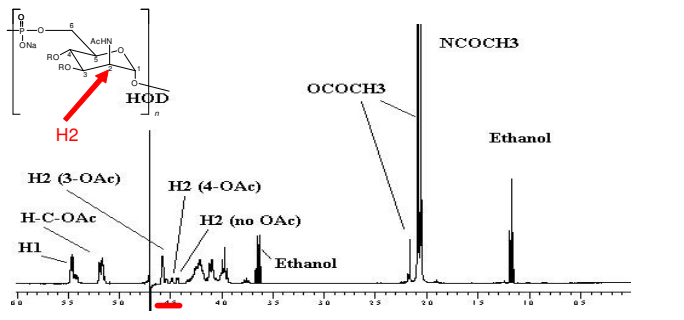

H2
HOD
NCOCH3
OCOCH3
Ethanol
H2 (3-OAc)
H2 (4-OAc)
H-C-OAc
H2 (no OAc)
H1
Ethanol

- **Characteristic proton profile (identity) and residuals (ethanol)**
- **Degree of acetylation from H1 and <u>H</u>-C-OAc (H3 and H4) or H2's [6]**

(>61.5%, calculated from ≥ 2 mmol acetyl/g polysaccharide [7]).

¹H NMR: Mn A PS structure/identity-2

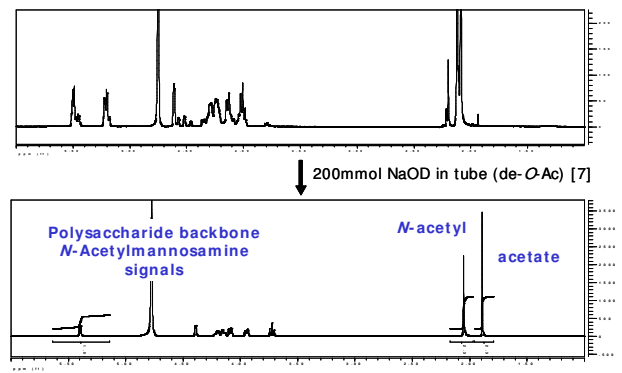

200mmol NaOD in tube (de-O-Ac) [7]
Polysaccharide backbone
N-Acetylmannosamine signals
N-acetyl
acetate

- **Identity of Mn A saccharide and O-acetylation -simplied**

¹H NMR: Tracking the PS activation (NaIO$_4$)

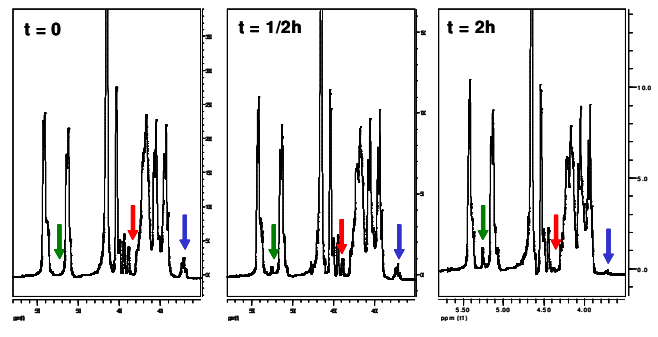

t = 0
t = 1/2h
t = 2h

- **Periodate oxidises the unacetylated residues**
- **Follow loss of H2 (↓) and H3 (↓) signals & appearance of new H1 (↓).**

¹H NMR: Tracking the conjugation process

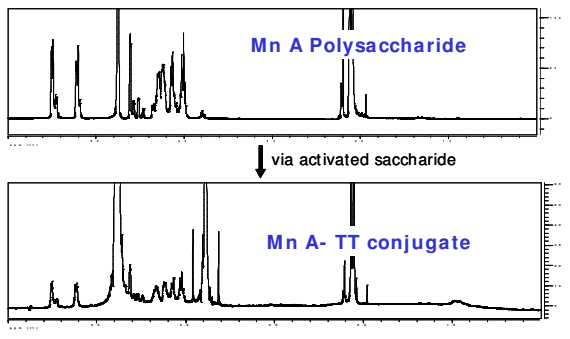

Mn A Polysaccharide
via activated saccharide
Mn A- TT conjugate

- **Provides evidence for maintenance of the structural integrity of Mn A saccharide through the process**

5. CONCLUSIONS

SEC-HPLC can be used to profile the size and heterogeneity of the polysaccharide, protein carrier, activated intermediates and conjugate vaccine, as well as to track the conjugation reaction.

The sensitive HPAEC-PAD technique can be used to quantify the saccharide throughout the process and to evaluate the integrity of the conjugate.

NMR spectroscopy provides a window into the structural integrity of the saccharide component (and *O*-acetylation) from polysaccharide to conjugate vaccine.

Fingerprinting by chromatography and NMR spectroscopy can be used to control the quality of conjugate vaccines and to demonstrate lot to lot manufacturing consistency

Phase I clinical trials completed (Poster P8.1.06)
Phase II in The Gambia and Mali (September 06)

6. REFERENCES

[1] MVP is a partnership between WHO and PATH with the goal of eliminating epidemic meningitis (group A) from sub-Saharan Africa through the development, testing, licensure and widespread use of conjugate meningococcal vaccines (www.meningvax.org/). [2] C. E. Frasch, S. Kapre, S. Beri, D. M. Granoff, N. Bouveret, F. M. LaForce FM, C.-H. R. Lee, A novel conjugation process for production of a highly immunogenic Group A meningococcal conjugate vaccine for use in Africa, Abstract, 14th International Pathogenic Neisseria Conference, Milwaukee, Wisconsin, USA. September 5-10, 2004. [3] Draft WHO Recommendations to assure the quality, safety and efficacy of group A meningococcal conjugate vaccines, to be presented to the Expert Committee on Biological Standardization (ECBS) in October, 2006. [4] Physico-Chemical Procedures for the Characterization of Vaccines, Dev. Biol. (Karger, 2000) F. Brown, M. Corbel, E. Griffiths Eds., vol. 103. [5] S. Ricci, A. Bardotti,S. D'Ascenzi, N. Ravenscroft, Development of a new method for the quantification of extracellular polysaccharide of *Neisseria meningitidis* serogroup A by high-performance anion-exchange chromatography with pulsed-amperometric detection, Vaccine, 2001, 19, 1989-1997. [6] Full ¹H NMR assignment and detailed O-acetylation patterns of capsular polysaccharides from *Neisseria meningitidis* used in vaccine production. X. Lemercinier, C. Jones, Carbohydr. Res. 1996, 296, 83 - 96. [7] Use and validation of NMR assays for the identity and O-acetyl content of capsular polysaccharides from Neisseria meningitidis used in vaccine manufacture, C. Jones, X. Lemercinier, J. Pharm. Biomed. Anal. 2002, 30, 1233 – 1247.

7. ACKNOWLEDGEMENTS

Financial support from the University of Cape Town, the National Research Foundation and the Ernest Oppenheimer Overseas Sabbatical Fellowship (for NR) is gratefully acknowledged. We thank friends and colleagues at the National Institute for Biological Standards and Control, UK (Drs Ian Feavers, Barbara Bolgiano and Chris Jones) for many valuable discussions and collaboration on the analysis of conjugates over the past few years. We also thank Xavier Lemercinier (NIBSC) for recording some of the NMR spectra presented here.